Aziza P. Manceur[1], Sonia Tremblay[1], Sven Ansorge[1]
[1] National Research Council, Montreal, Quebec, H4P 2R2, Canada

NRC·CNRC

Human Health Therapeutics, Vaccine Program

Insulin increases influenza virus yield in HEK293 cells for vaccine production

OBJECTIVES

1. **Evaluate the effect of insulin on HEK293SF-3F6[1] cell growth in two types of media.**
2. **Boost influenza production with addition of different concentrations of insulin.**
3. **Quantify influenza using a robust method.**

1) EFFECT OF INSULIN ON CELL GROWTH

- HEK293SF-3F6[1] is a suspension-adapted cell line (a GMP cell bank is available).
- HEK293SF-3F6[1] cells have successfully been used for the production of influenza virus, virus-like particles, lentiviral vectors, adenovirus and adeno-associated virus.
- HEK293SF-3F6[1] can be cultured in an in-house media (IHM-03) and in commercially-available serum-free media (Media 1) but weak growth profiles are obtained in chemically-defined media.

1.1) Growth limitations observed in a chemically-defined media (Media 1) were alleviated by insulin:

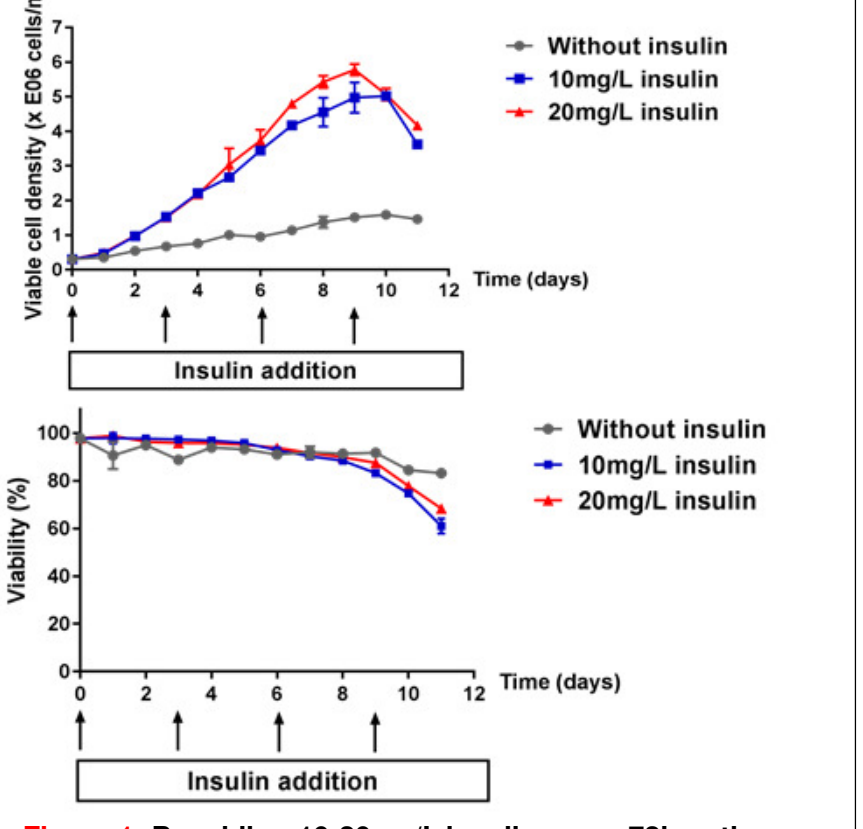
Viable cell density (x E06 cells/ml)
Time (days)
Without insulin
10mg/L insulin
20mg/L insulin
Insulin addition
Viability (%)
Time (days)
Without insulin
10mg/L insulin
20mg/L insulin
Insulin addition

Figure 1: By adding 10-20mg/L insulin every 72hrs, the maximal viable cell density of HEK293SF-3F6 cells was increased by 4-fold in Media 1.

1.2) Growth kinetics were improved by insulin in an In-House Media (IHM-03):

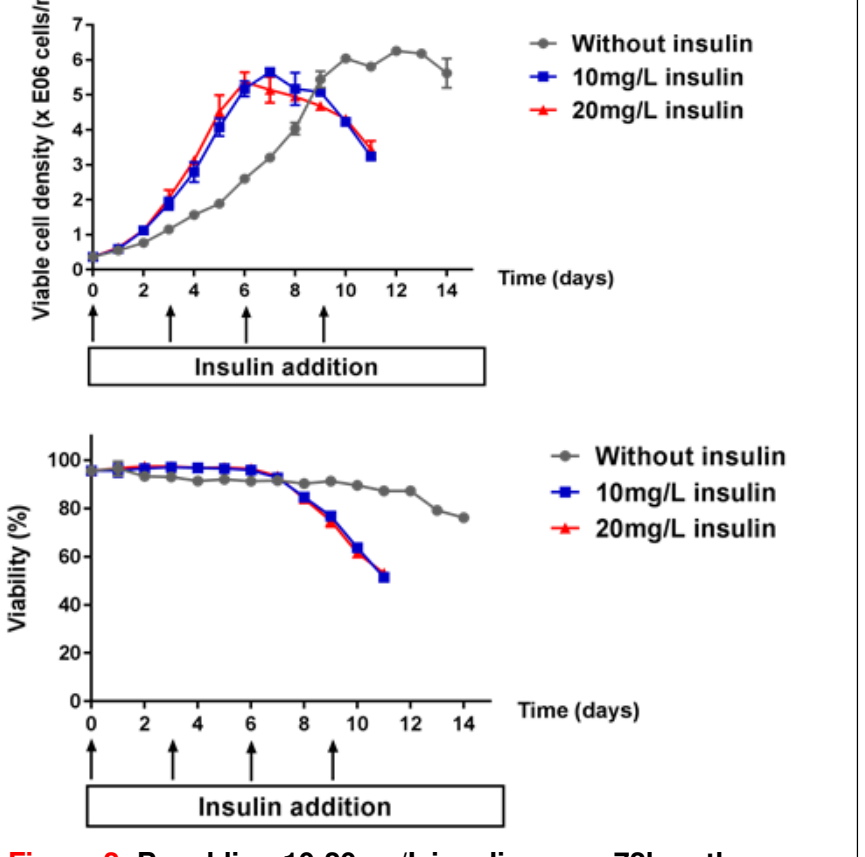
Viable cell density (x E06 cells/ml)
Time (days)
Without insulin
10mg/L insulin
20mg/L insulin
Insulin addition
Viability (%)
Time (days)
Without insulin
10mg/L insulin
20mg/L insulin
Insulin addition

Figure 2: By adding 10-20mg/L insulin every 72hrs, the maximal viable cell density of HEK293SF-3F6 cells was reached 3 days sooner in IHM-03, a media designed specifically for HEK293SF-3F6 cells.

2) EFFECT OF INSULIN ON INFLUENZA PRODUCTION IN HEK293SF-3F6 CELLS

- Cell-culture based vaccines are a valuable alternative to egg-produced vaccines: the equivalent of 1500 influenza vaccine doses can be produced in a 1L bioreactor within 48 hours with HEK293 cells.

2.1) Insulin is a strong activator of the PI3K/Akt pathway, which plays a key role in influenza production:

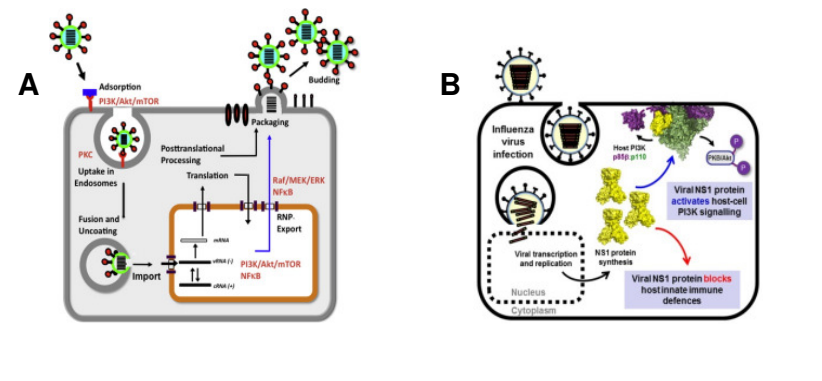
A
Adsorption
PI3K/Akt/mTOR
Budding
Packaging
PKC
Uptake in Endosomes
Posttranslational Processing
Translation
Raf/MEK/ERK
NFκB
RNP Export
Fusion and Uncoating
Import
PI3K/Akt/mTOR
NFκB
B
Influenza virus infection
Viral NS1 protein activates host-cell PI3K signalling
Viral transcription and replication
NS1 protein synthesis
Viral NS1 protein blocks host innate immune defences
Nucleus
Cytoplasm

Figure 3. A) Overview of signaling pathways modulated by influenza infection: The replication of influenza virus hijacks the cellular machinery and involves multiple signaling pathways including Akt[1]. B) Akt is activated by viral NS-1[2].

2.2) Insulin increases the yield of influenza virus in a 24-well microbioreactor:

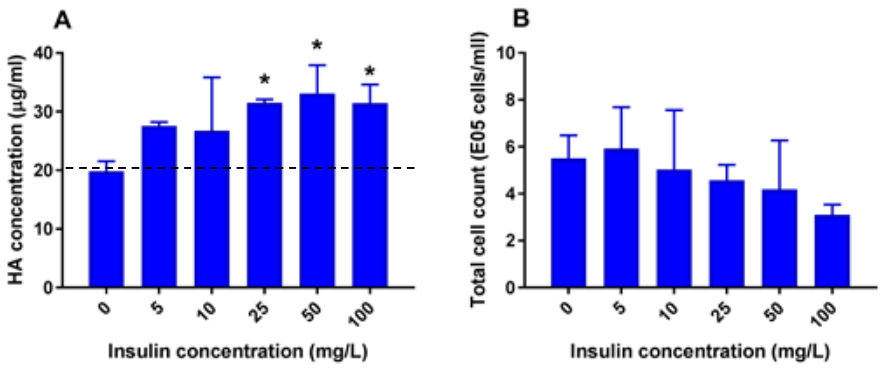
A
HA concentration (µg/ml)
Insulin concentration (mg/L)
B
Total cell count (E05 cells/ml)
Insulin concentration (mg/L)

Figure 4. Addition of 25-100 mg/L insulin at time of infection increases the yield of influenza H1N1 A/Puerto Rico 8/34 by almost 2-fold in Media 1 (A), without affecting the total cell count at harvest (B). The virus was added at an MOI of 0.01 at 35oC in the presence of 1µg/ml trypsin-TPCK. HA concentration was measured by dot-blot using pan-HA antibodies developed in-house (See panel 3).

- Increase in total HA was not due to increased cell density
- Similar results were obtained with H3N2 A/Aichi/2/68 treated with 5 and 25 mg/L insulin at time of infection

2.3) Akt activity is increased by influenza infection and insulin addition in HEK293SF-3F6 cells:

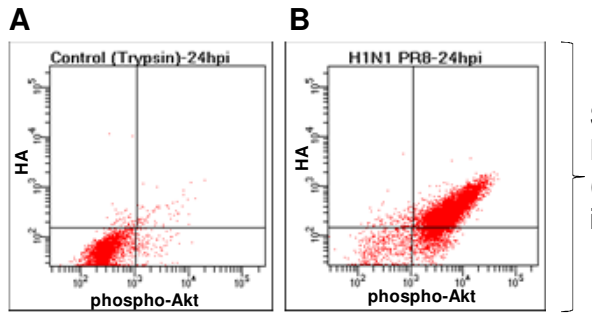
A
Control (Trypsin)-24hpi
HA
phospho-Akt
B
H1N1 PR8-24hpi
HA
phospho-Akt

Strong Akt activation in HA-positive population (cells infected by influenza) after 24hrs

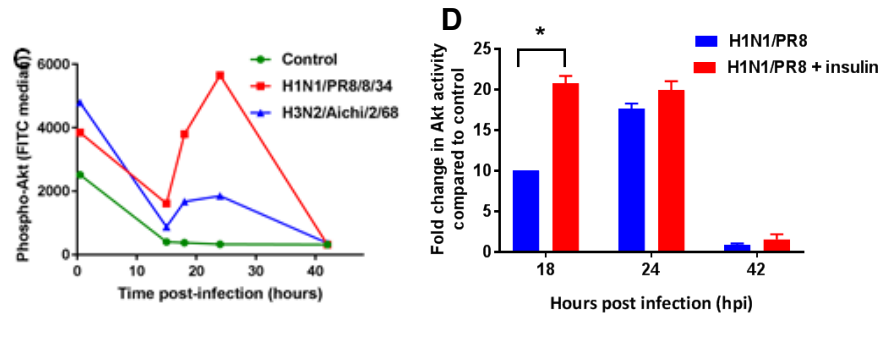
C
Phospho-Akt (FITC median)
Time post-infection (hours)
Control
H1N1/PR8/8/34
H3N2/Aichi/2/68
D
Fold change in Akt activity compared to control
Hours post infection (hpi)
H1N1/PR8
H1N1/PR8 + insulin

Figure 5. HEK293 cells were infected in a shake flask. The control was treated with 1 µg/ml trypsin-TPCK (without virus). Phospho-Akt was measured by flow cytometry after infection and representative results are shown (A-B) and summarized (C). The addition of insulin further activates Akt (D). Results were obtained by dividing the fluorescence signal obtained for the infected sample by the untreated control (n=2).

3) INFLUENZA CAN BE QUANTIFIED WITH PAN-HA ANTIBODIES IN A DOT-BLOT ASSAY

- Quantification of influenza is an enduring challenge:
 - Measures of infectivity are highly variable
 - Physical methods to count viral particles can be confounded by the presence of non-viral particles
 - Antibody-dependent methods to quantify surface proteins such as hemagglutinin (HA) rely on strain-specific antibodies

→ A pan-HA antibody cocktail was generated using a highly conserved peptide sequence found within the HA molecule (the fusion peptide)

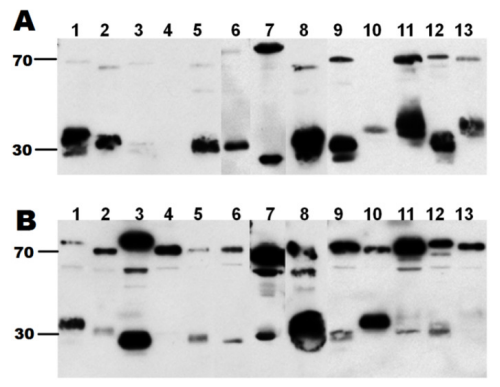
A
1 2 3 4 5 6 7 8 9 10 11 12 13
70
30
B
1 2 3 4 5 6 7 8 9 10 11 12 13
70
30

Lane	Subtype	Strain
1	A/H1N1	A/Puerto Rico/8/34
2	A/H2N2	A/Singapore/1/57
3	A/H3N2	A/New York/55/01
4	A/H4N6	A/Duck/Czechoslovakia/56
5	A/H5N9	A/Turkey/Wisconsin/68
6	A/H6N2	A/Turkey/Massachusetts/3740/65
7	A/H7N7	A/Equine/Prague/1/56
8	A/H8N4	A/Turkey/Ontario/6118/68
9	A/H9N2	A/Turkey/Wisconsin/1/66
10	A/H10N8	A/Quail/Italy/1117/65
11	A/H11N6	A/Duck/England/56
12	A/H12N6	A/Duck/Wisconsin/480/79
13	A/H13N6	A/Gull/Maryland/704/77

Figure 6. Western blots of influenza strains belonging to 13 different subtypes. Influenza viruses were produced in eggs and probed with mAb 11H12 (A) or mAb 10A9 (B). When mixed together, the two mAb make a pan-HA cocktail.

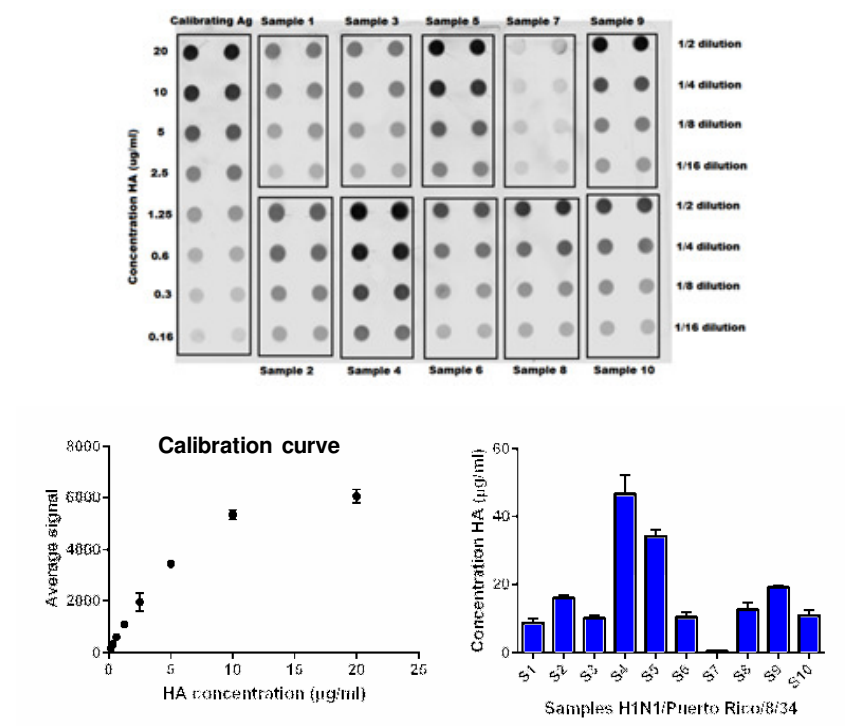
Calibrating Ag
Sample 1
Sample 3
Sample 5
Sample 7
Sample 9
Sample 2
Sample 4
Sample 6
Sample 8
Sample 10
Concentration HA (ug/ml)
1/2 dilution
1/4 dilution
1/8 dilution
1/16 dilution
Calibration curve
Average signal
HA concentration (µg/ml)
Concentration HA (µg/ml)
Samples H1N1/Puerto Rico/8/34

Figure 7. Quantification by dot blot. A calibration curve ranging from 160ng/ml to 20µg/ml HA was obtained using a calibrating antigen of known concentration. Ten different samples produced in HEK293SF-3F6 cells were loaded in 4 dilutions and the concentration of HA was determined. Samples S4 and S5 were purified by sucrose-cushion while the other samples were crude samples harvested after different treatments.

5) CONCLUSION

- The HEK293SF-3F6 cell line platform is an ideal candidate for the development of commercial manufacturing processes of virus and viral vectors for vaccines and gene/cell therapy.
- Influenza concentration in supernatant was measured using pan-HA antibodies produced in-house using a peptide-conjugate derived from the fusion peptide.
- Overall, a concentration of 25mg/L insulin provided an increase in influenza yield regardless of the media or viral strain used in HEK293SF-3F6 cells.
- A concomitant activation of signaling pathways associated with cell survival (PI3K-Akt pathway) was observed.

References and Acknowledgements

1. Proprietary of NRC under Canadian Patent Document number 2252972 and US Patent: number 6,210,922, available for commercial licensing
2. Planz O., Antiviral Research, Volume 98, issue 3, June 2013
3. http://www.virology.uzh.ch/research/ghale.html

Thank you to Magnus Franzmann and Clare Medlow (Novo Nordisk Pharmatech) for scientific input and for providing insulin.

The results described do not represent an endorsement of any product by NRC.

National Research Council Canada
Conseil national de recherches Canada
Canada